Role of selectins in the pathogenesis of multiple myeloma

A.K. Azab[1], F. Azab[1], J.M. Runnels[1], A. M. Roccaro[1], J. L. Magnani[3], A. Sarkar[3], K. C. Anderson[1], C. P. Lin[2], I. M. Ghobrial[1]

[1]Medical Oncology, Dana-Farber Cancer Institute, [2]Wellman Center for Photomedicine, Massachusetts General Hospital; Harvard Medical School, Boston, MA, USA. [3]GlycoMimetics Inc, Gaithersburg, MD, USA

Introduction

The interaction of multiple myeloma (MM) cells with extracellular matrix (ECM) proteins, bone marrow (BM) stromal cells (BMSCs) and chemokines in the BM milieu plays a crucial role in MM pathogenesis and drug resistance. These molecular events are triggered either by MM cell adhesion to BMSCs and ECM; or by chemokines. We have previously shown that the chemokine stromal cell-derived factor-1 (SDF-1) and its receptor, CXCR4, regulate chemotaxis and homing of MM cells to the BM. Moreover, SDF-1 was shown to induce MM proliferation, upregulate VLA-4-mediated cell adhesion to both fibronectin and VCAM-1, and increase chemotaxis, invasion and actin polymerization in MM cells.

Selectins are adhesion molecules expressed by activated endothelium of venules and leukocytes, and are involved in the primary interaction of lymphocytes with the endothelium of blood vessels. The binding of selectins serves as a biologic brake, making the leukocyte quickly decelerate by rolling on endothelial cells, as the first step of extravasation. In this study, we will focus on the role of selectins and their ligands in the regulation of homing of MM cells to the BM and the therapeutic implications of this role.

Results

Figure 1: Expression of selectin-ligands in MM patient samples, cell lines and normal plasma cells

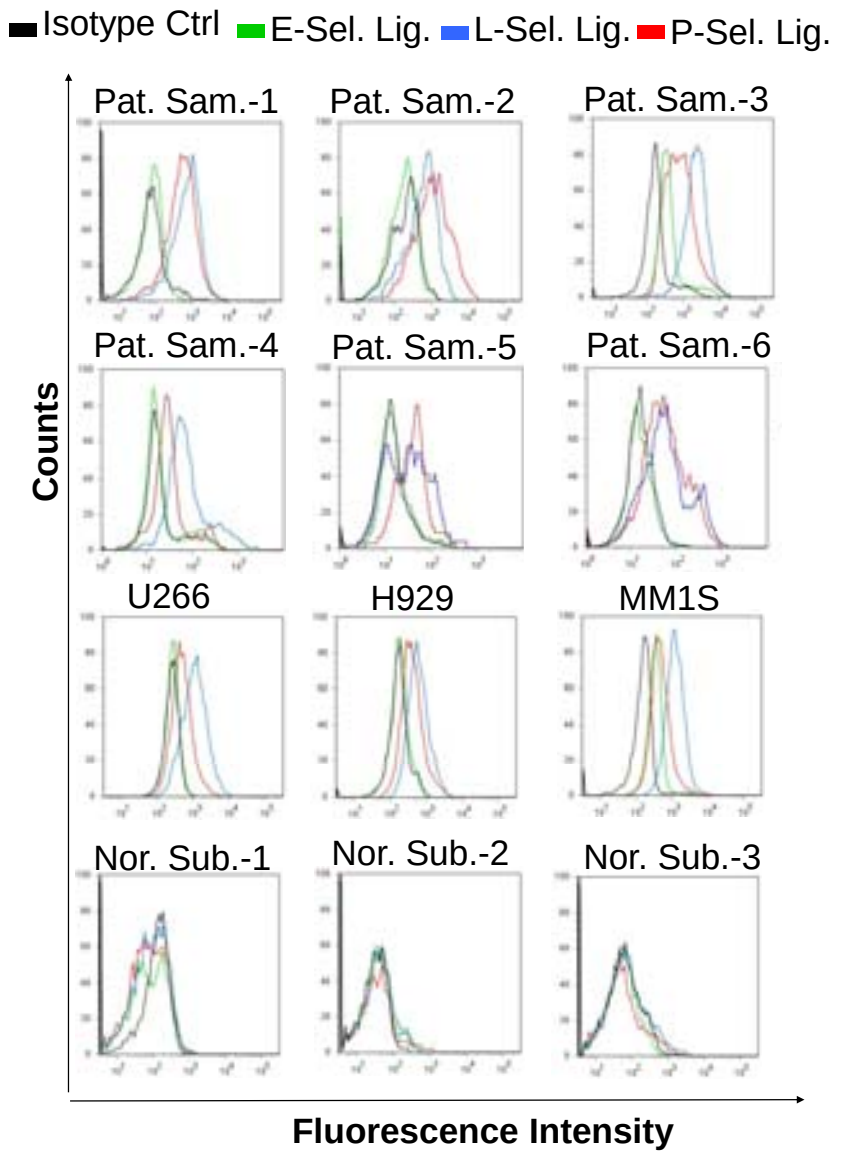
Isotype Ctrl
E-Sel. Lig.
L-Sel. Lig.
P-Sel. Lig.
Pat. Sam.-1
Pat. Sam.-2
Pat. Sam.-3
Pat. Sam.-4
Pat. Sam.-5
Pat. Sam.-6
U266
H929
MM1S
Nor. Sub.-1
Nor. Sub.-2
Nor. Sub.-3
Counts
Fluorescence Intensity

Figure 2: The role of selectins in Adhesion of MM cell to HUVEC cells.

The effect of treating HUVEC cells with anti-antibodies against selectin

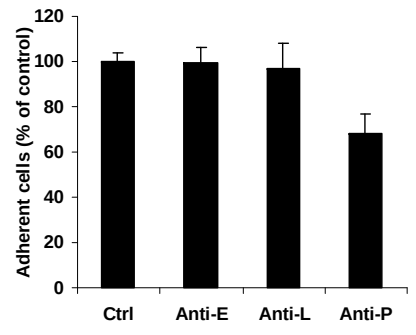
Adherent cells (% of control)
Ctrl
Anti-E
Anti-L
Anti-P

The effect of treating MM cells with recombinant selectin

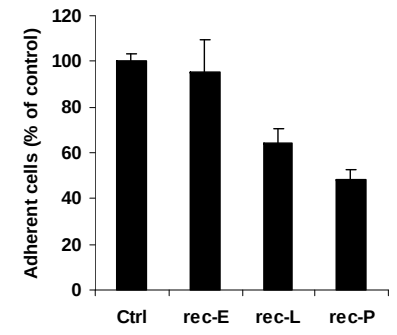
Adherent cells (% of control)
Ctrl
rec-E
rec-L
rec-P

The effect of activating HUVEC cells to express E and P selectin.

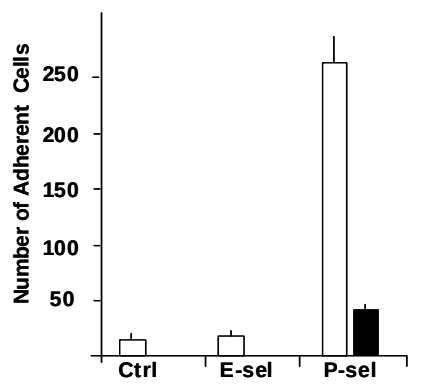
Number of Adherent Cells
Ctrl
E-sel
P-sel

The effect of treating HUVEC cells with GMI-1070

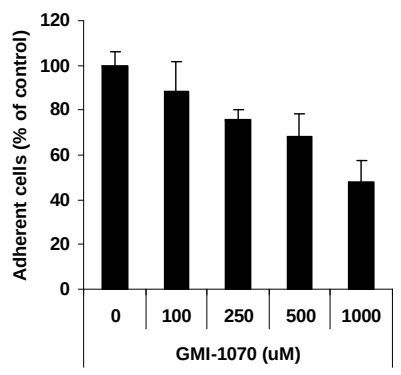
Adherent cells (% of control)
0
100
250
500
1000
GMI-1070 (uM)

Figure 3: The role of selectins in chemotaxis of MM cell in response to SDF1.

The effect of GMI-1070 on trans-well migration

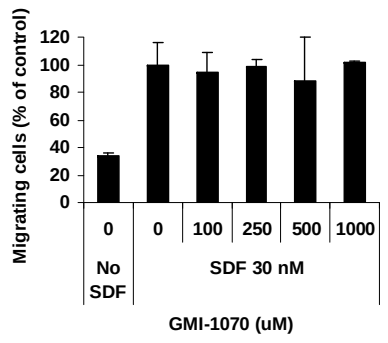
Migrating cells (% of control)
No SDF
SDF 30 nM
GMI-1070 (uM)

The effect of GMI-1070 on trans-endothelial migration

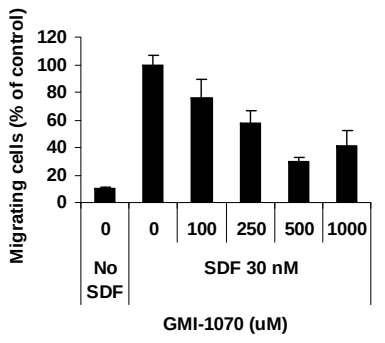
Migrating cells (% of control)
No SDF
SDF 30 nM
GMI-1070 (uM)

The effect of SDF on the expression level of selectins in HUVEC cells

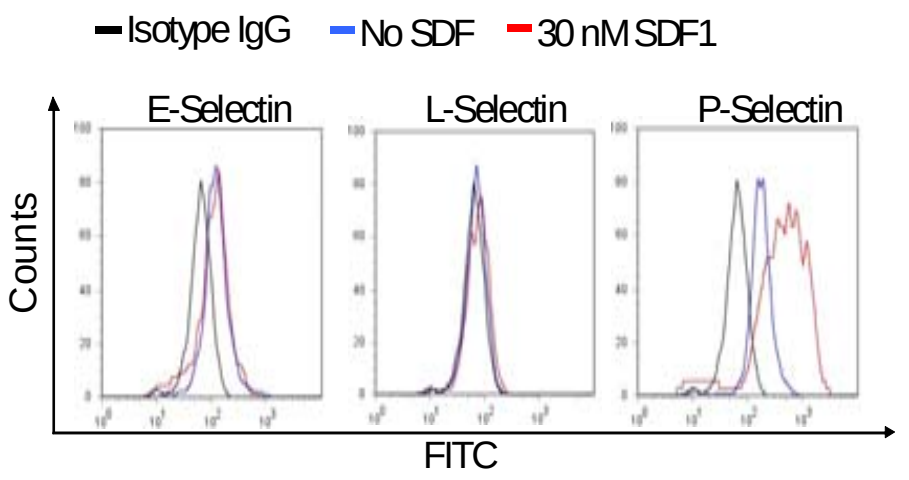
Isotype IgG
No SDF
30 nM SDF1
E-Selectin
L-Selectin
P-Selectin
Counts
FITC

Figure 4: The effect of GMI-1070 on the circulation time of MM cells after iv injection in mice.

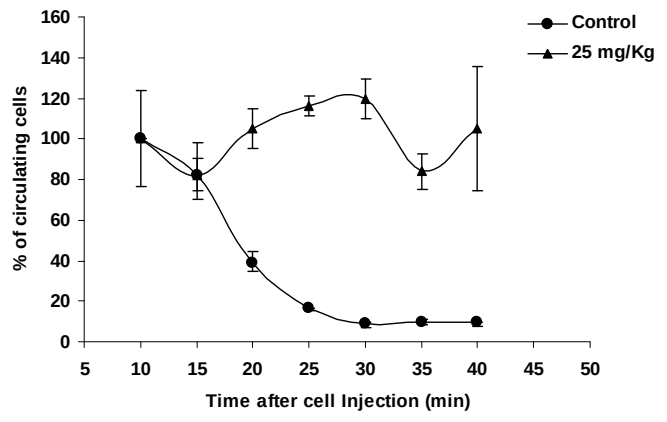
Control
25 mg/Kg
% of circulating cells
Time after cell Injection (min)

Figure 5: Selectin signaling in MM

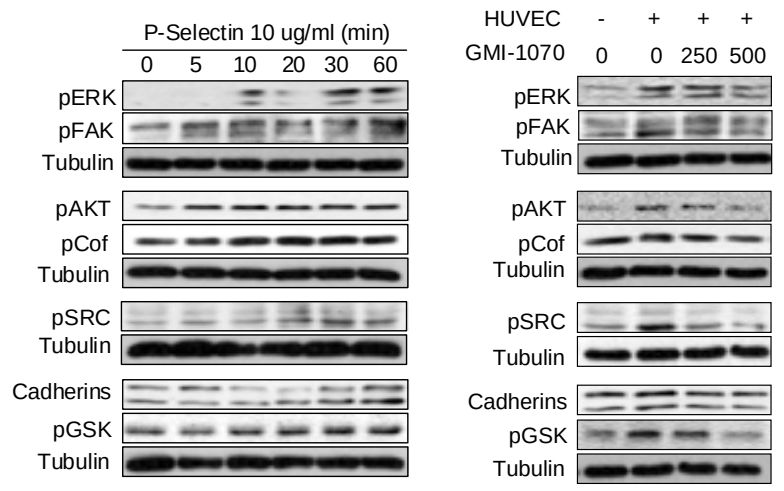
P-Selectin 10 ug/ml (min)
0 5 10 20 30 60
HUVEC - + + +
GMI-1070 0 0 250 500
pERK
pFAK
Tubulin
pAKT
pCof
pSRC
Cadherins
pGSK

Conclusion

We found that L and P selectin ligands are highly expressed in MM cells compared to normal plasma cells. Mainly P-selecin played a major role in homing of MM cells to the BM. Moreover, the pan-selectin inhibitor GMI-1070 prevented extravasation and thereby the homing of MM cells to the BM and prevented signaling induced by HUVEC. This provides a basis for testing the effect of the inhibitor on MM tumor progression and initiation.

Methods

We have characterized the expression of E, L and P-selectins and their ligands on MM cell lines, patient samples and plasma cells from normal subjects. We have tested the effect of blockade of each of the selectins and selectin-ligands on the interaction of MMCs with ECs. Moreover, we tested the effect of a pan selectin inhibitor on MMCs adhesion to ECs, and trans-well (through filter) and trans-endothelial SDF1-induced migration in vitro, and characterized its effect on cytoskeletal signaling induced by the interaction of MMCs and ECs. Moreover, we have tested the effect of the inhibitor on homing of MMCs to the BM in mice using in vivo flow cytometry to detect the number of circulating cells.

Acknowledgments

Supported by: MMRF; NIH R01 CA125690; and Leukemia and Lymphoma Society; and ASH scholar grant.

Corresponding:
Kareem Azab, B.Pharm. Ph.D.
Harvard Medical School, Dana-Farber Cancer Institute,
44 Binney Street, Dana 838, Boston, MA, 02115.
Phone: (617)-632-2944,Fax: (617)-632-8608
Email: abdelkareem_azab@dfci.harvard.edu