Trials in Progress: A Phase 3 Trial to Evaluate the Efficacy of Uproleselan (GMI-1271) with Chemotherapy In Patients with Relapsed/Refractory Acute Myeloid Leukemia

Daniel J. DeAngelo, MD, PhD[1], Harry Erba, MD, PhD[2], Brian A. Jonas, MD, PhD[3], Michael E. O'Dwyer, MD[4], Paula Marlton, MD[5], Gerwin Huls, MD, PhD[6], Jane L. Liesveld, MD[7], Brenda Cooper, MD[8], Bhavana Bhatnagar, DO[9], Michael Armstrong, MD, PhD[10], William Fogler, PhD[11], Mary Chen, MD, PhD[11], John Magnani, PhD[11], Helen Thackray, MD[11], Eric Feldman, MD[11], Anjali S. Advani, MD[12], and Pamela S. Becker, MD[13]

[1]Department of Medical Oncology, Dana-Farber Cancer Institute, Boston, MA, [2]Department of Medicine, Duke University School of Medicine, Durham, NC, [3]Department of Medicine, Division of Hematology and Oncology, UC Davis Comprehensive Cancer Center, Sacramento, CA, [4]Department of Haematology, University Hospital Galway, Galway, Ireland, [5]Department of Haematology, Princess Alexandria Hospital, Queensland, Australia, [6]Department of Hematology, University Medical Center, Groningen, Netherlands, [7]Department of Medicine, University of Rochester Medical Center, School of Medicine and Dentistry, Rochester, NY, [8]Division of Hematology/Oncology, University Hospitals Cleveland Medical Center, Cleveland, OH, [9]Division of Hematology, The Ohio State University Wexner Medical Center, Columbus, OH, [10]IQVIA Biotech, Morrisville, NC, [11]GlycoMimetics, Inc., Rockville, MD, [12]Cleveland Clinic Taussig Cancer Insititute, Cleveland, OH, [13]University of Washington and Fred Hutchinson Cancer Research Center, Seattle, WA.

Background

Uproleselan (GMI-1271), an E-selectin antagonist, disrupts the relationship between tumor cells and bone marrow microenvironment (Figure 1)

Figure 1: Mechanism of Action

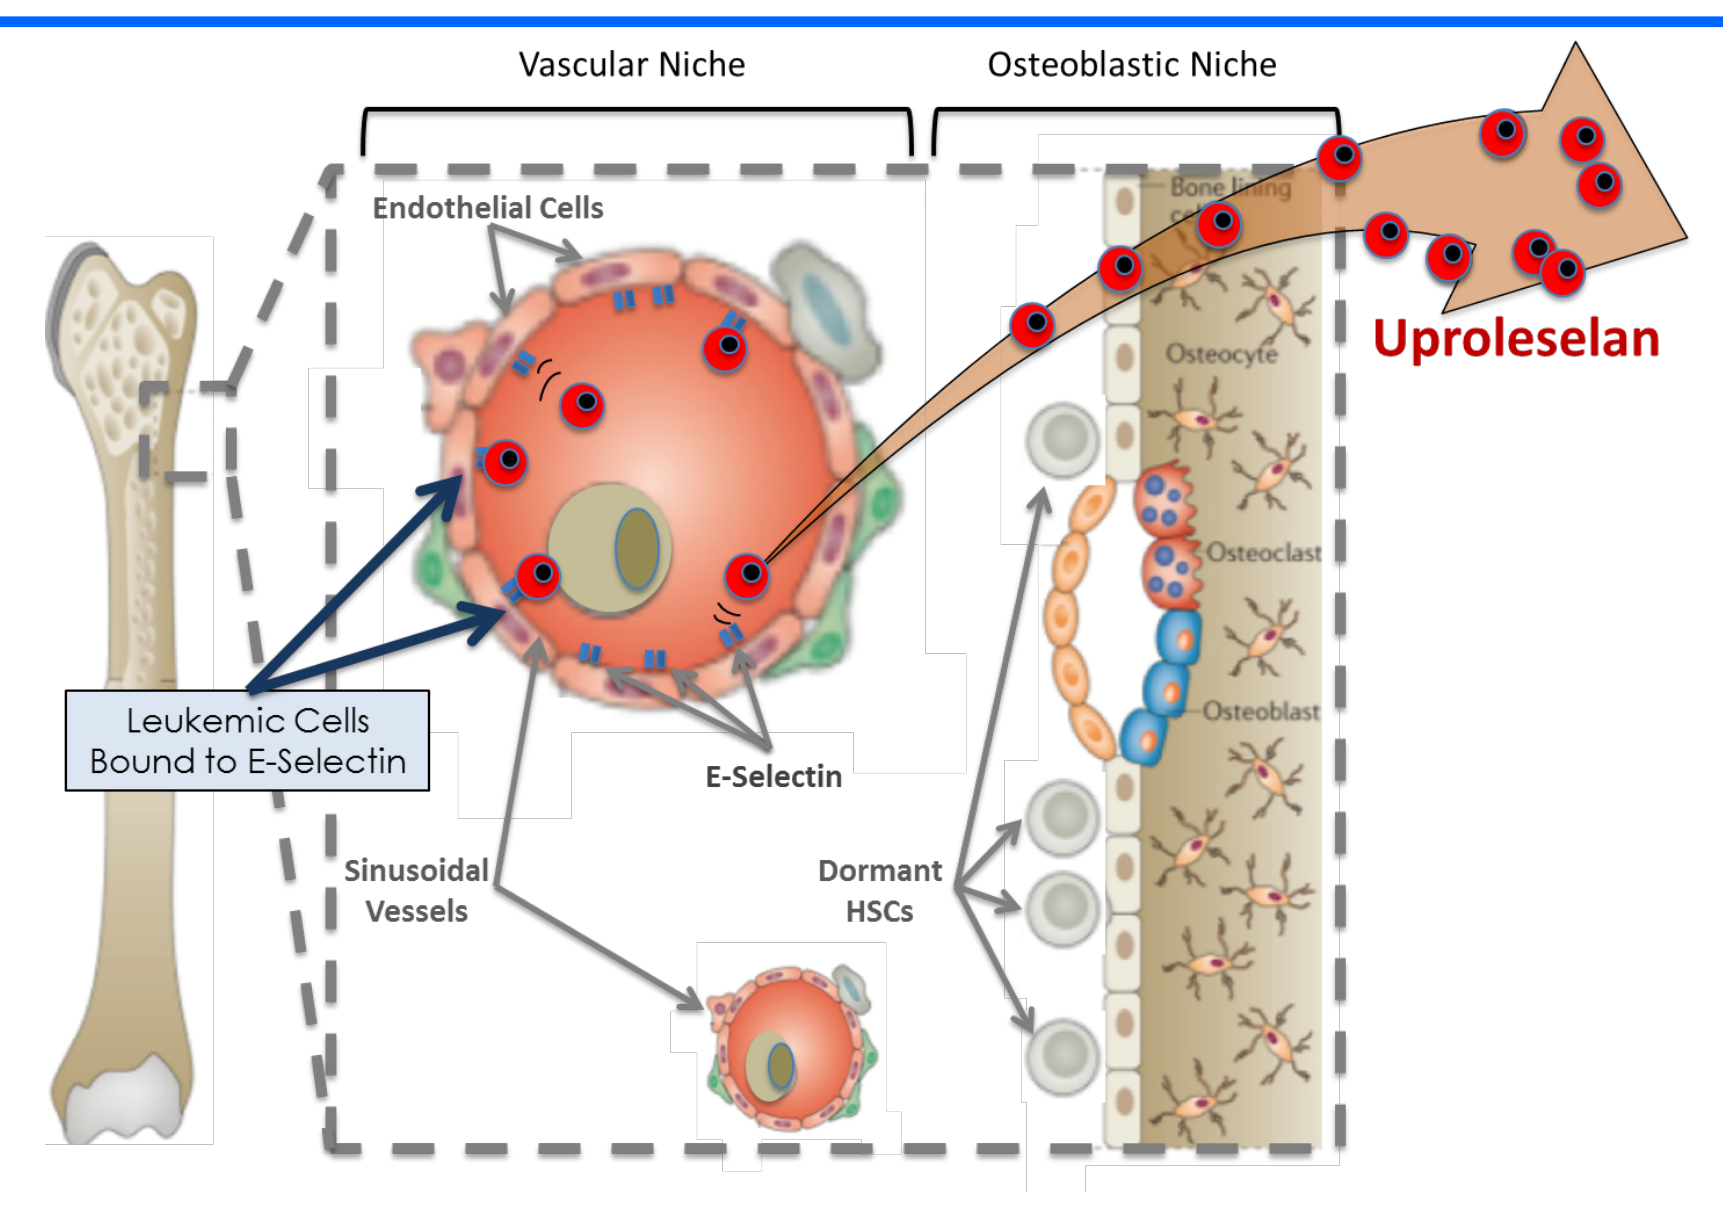

Vascular Niche
Osteoblastic Niche
Endothelial Cells
Uproleselan
Osteocyte
Osteoclast
Osteoblast
Leukemic Cells Bound to E-Selectin
E-Selectin
Sinusoidal Vessels
Dormant HSCs

Figure 2a: Uproleselan Protects Against Chemotherapy-Induced Mucosal Injury in Mice

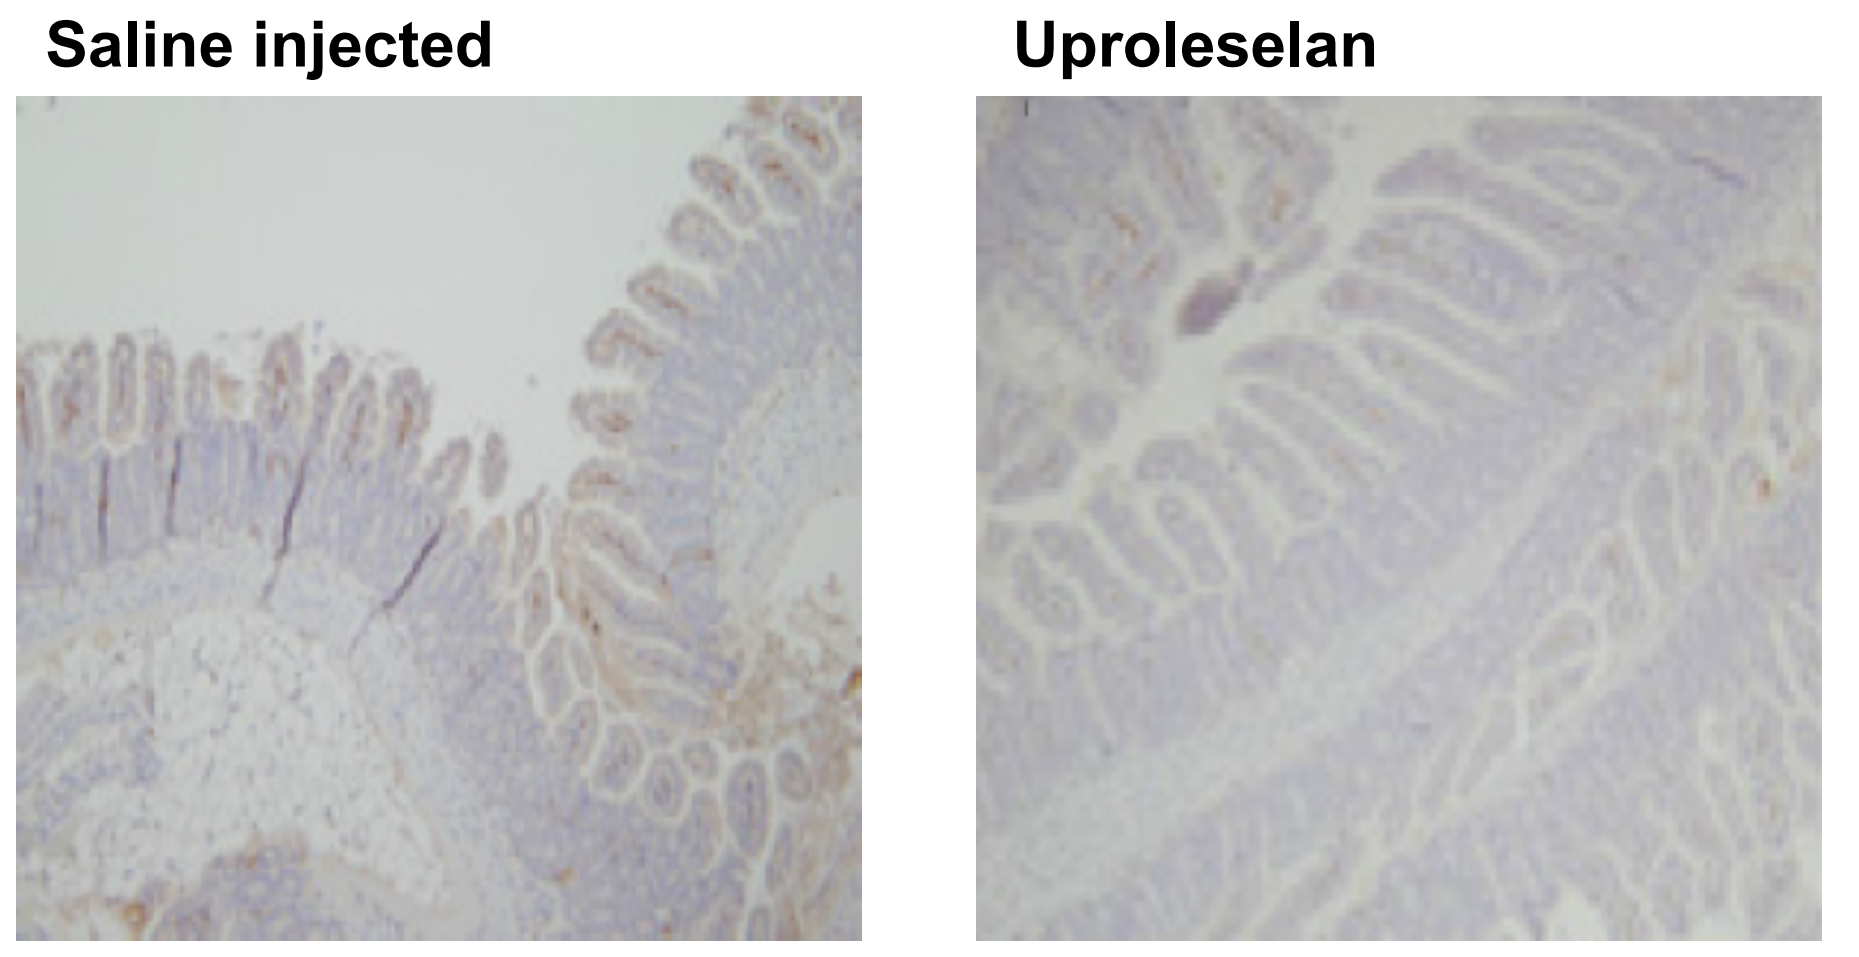

Saline injected
Uproleselan

Winkler, et al. 2014

Figure 2b: Uproleselan in Combination with Chemotherapy Prolongs Survival in AML Tumor Model

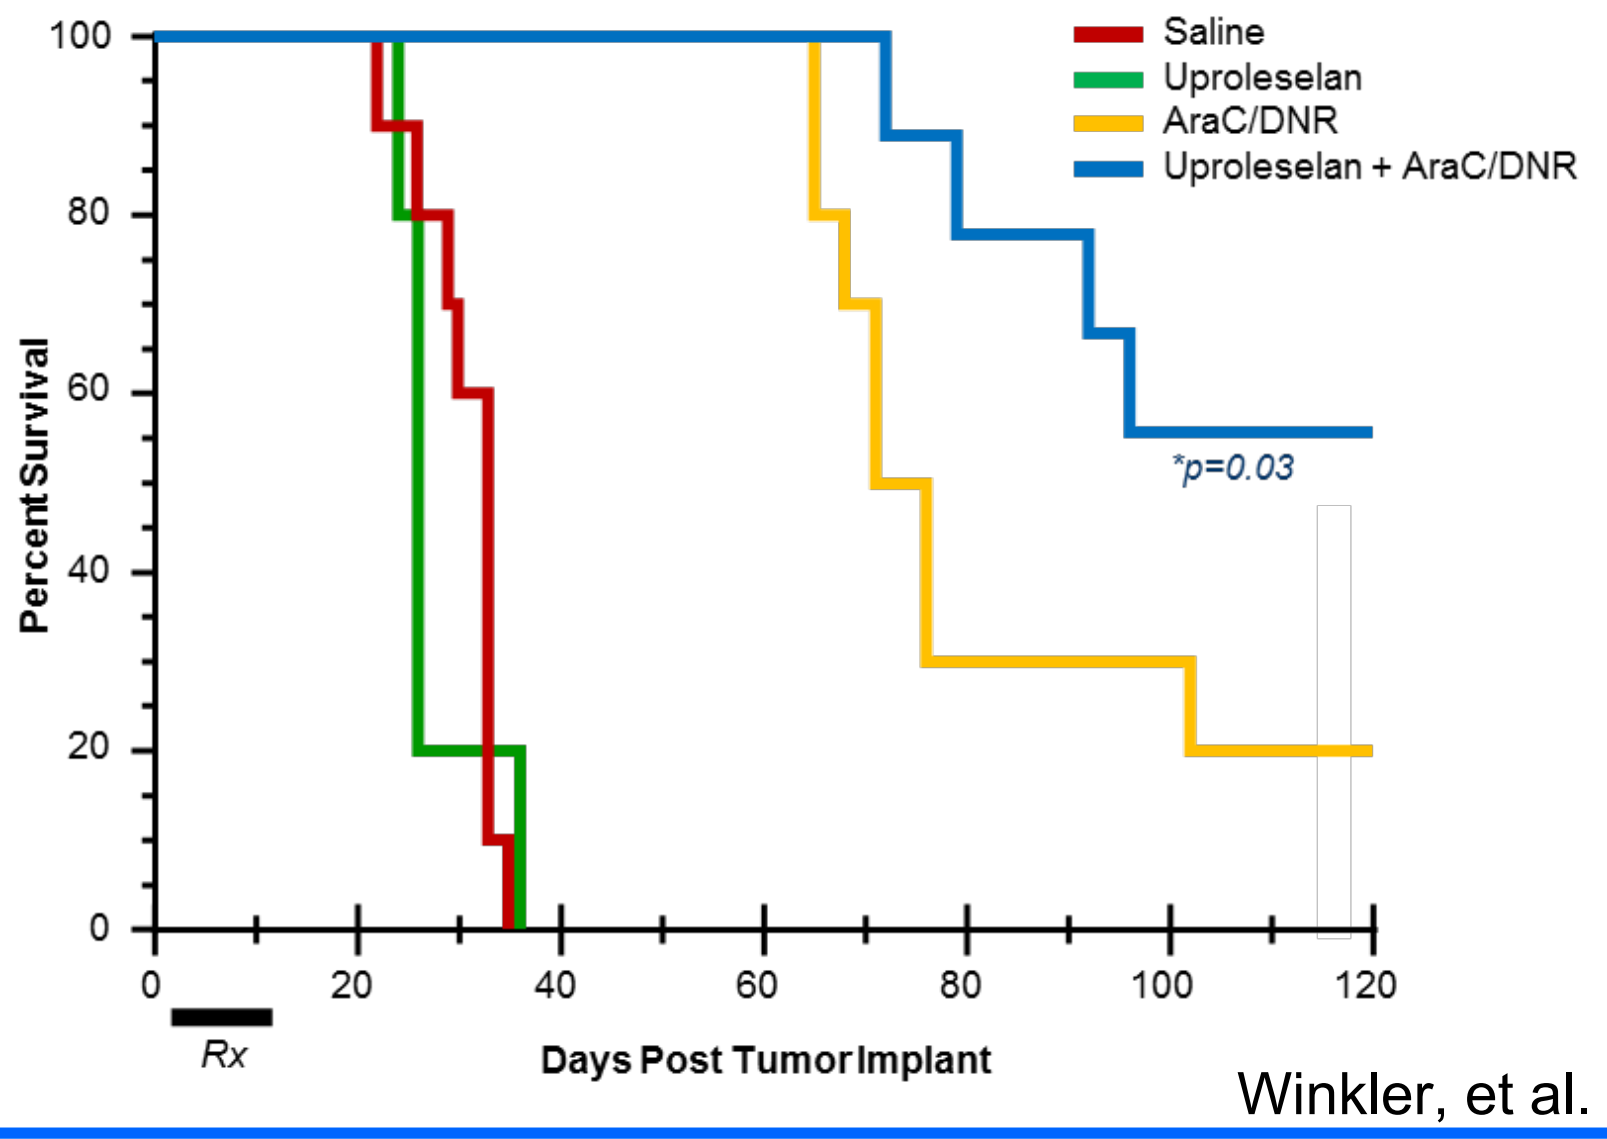

Saline
Uproleselan
AraC/DNR
Uproleselan + AraC/DNR
*p=0.03
Percent Survival
Rx
Days Post Tumor Implant

Winkler, et al. 2016

E-selectin
- Constitutively expressed in the bone marrow microvasculature
- Binds to the E-selectin ligands on AML cells
- Promotes environment-medicated drug resistance (EMDR) of leukemic cell

Uproleselan, an E-selectin antagonist:
- Inhibits activation of cancer survival pathways (e.g., NF-KB), disrupting EMDR within bone marrow
- Reduces chemotherapy-associated mucositis (Figure 2a)
- Prolongs survival over chemotherapy alone in animal models (Figure 2b)
- Protects normal HSCs by enhancing quiescence and ability for self-renewal

Key Findings in Phase I/II Trial

Phase I/II AML trial in relapsed/refractory (R/R) AML (uproleselan + MEC) and newly diagnosed older AML (uproleselan + '7+3') was completed.
- Key findings in R/R population:
 - Higher remission rate (CR/CRi) of 39% (historical control 28%, Feldman, et al. 2005)
 - High rates of MRD negativity
 - Improved overall survival (OS) of 8.8 months (historical control 5.2 months, Feldman, et al. 2005) (Figure 4a)
 - High E-sel-L expression on leukemic blasts correlated with improved OS (Figures 3 and 4b)
 - Low rates of severe oral mucositis
- Breakthrough Therapy Designation granted by FDA for R/R population (NCT03616470)
- Additional trials planned for front-line older newly diagnosed FIT/UNFIT AML (NCT03701308 and EudraCT 2018-003480-64)

Figure 3: E-selectin Ligand Detectable on Blasts in All Patients

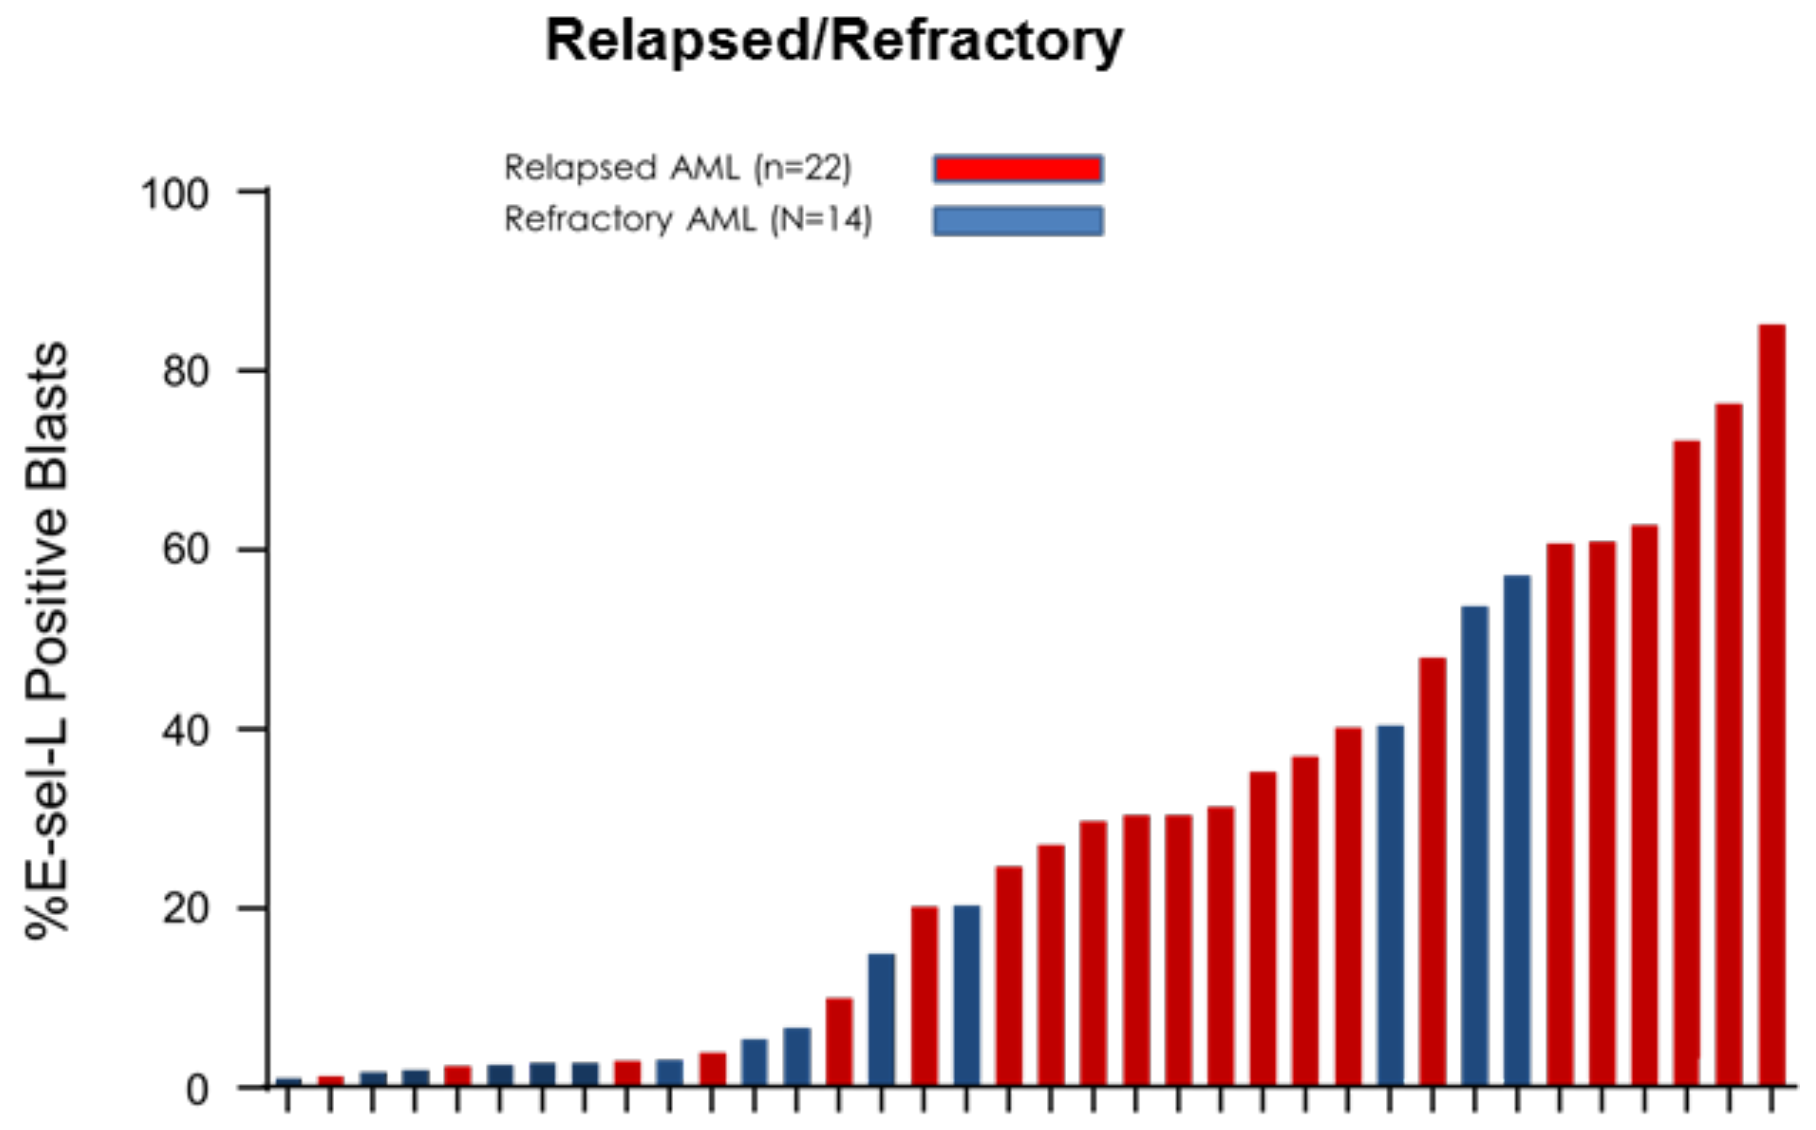

Relapsed/Refractory
Relapsed AML (n=22)
Refractory AML (N=14)
%E-sel-L Positive Blasts

Figure 4a: Overall Survival in All Patients Receiving RP2D

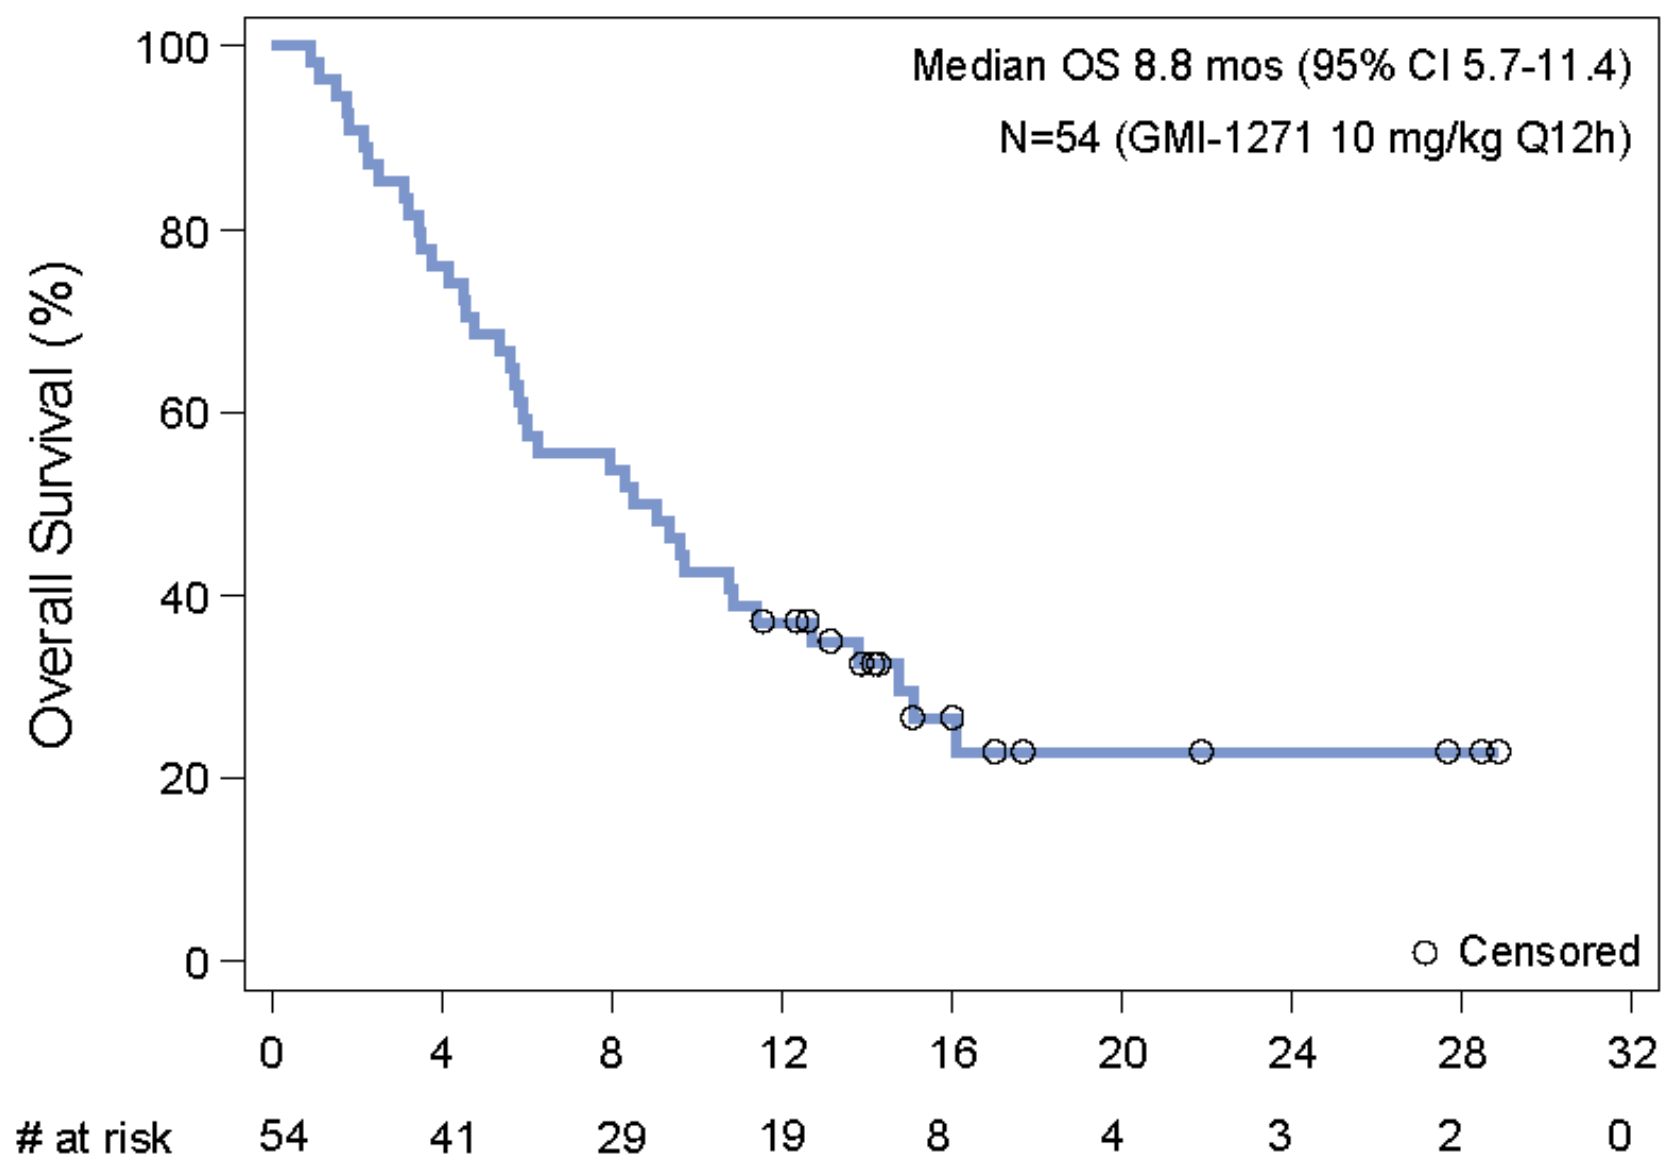

Median OS 8.8 mos (95% CI 5.7-11.4)
N=54 (GMI-1271 10 mg/kg Q12h)
Overall Survival (%)
Censored
at risk 54 41 29 19 8 4 3 2 0

Figure 4b: Overall Survival in High E-sel-L Expression vs. Low E-sel-L Expression

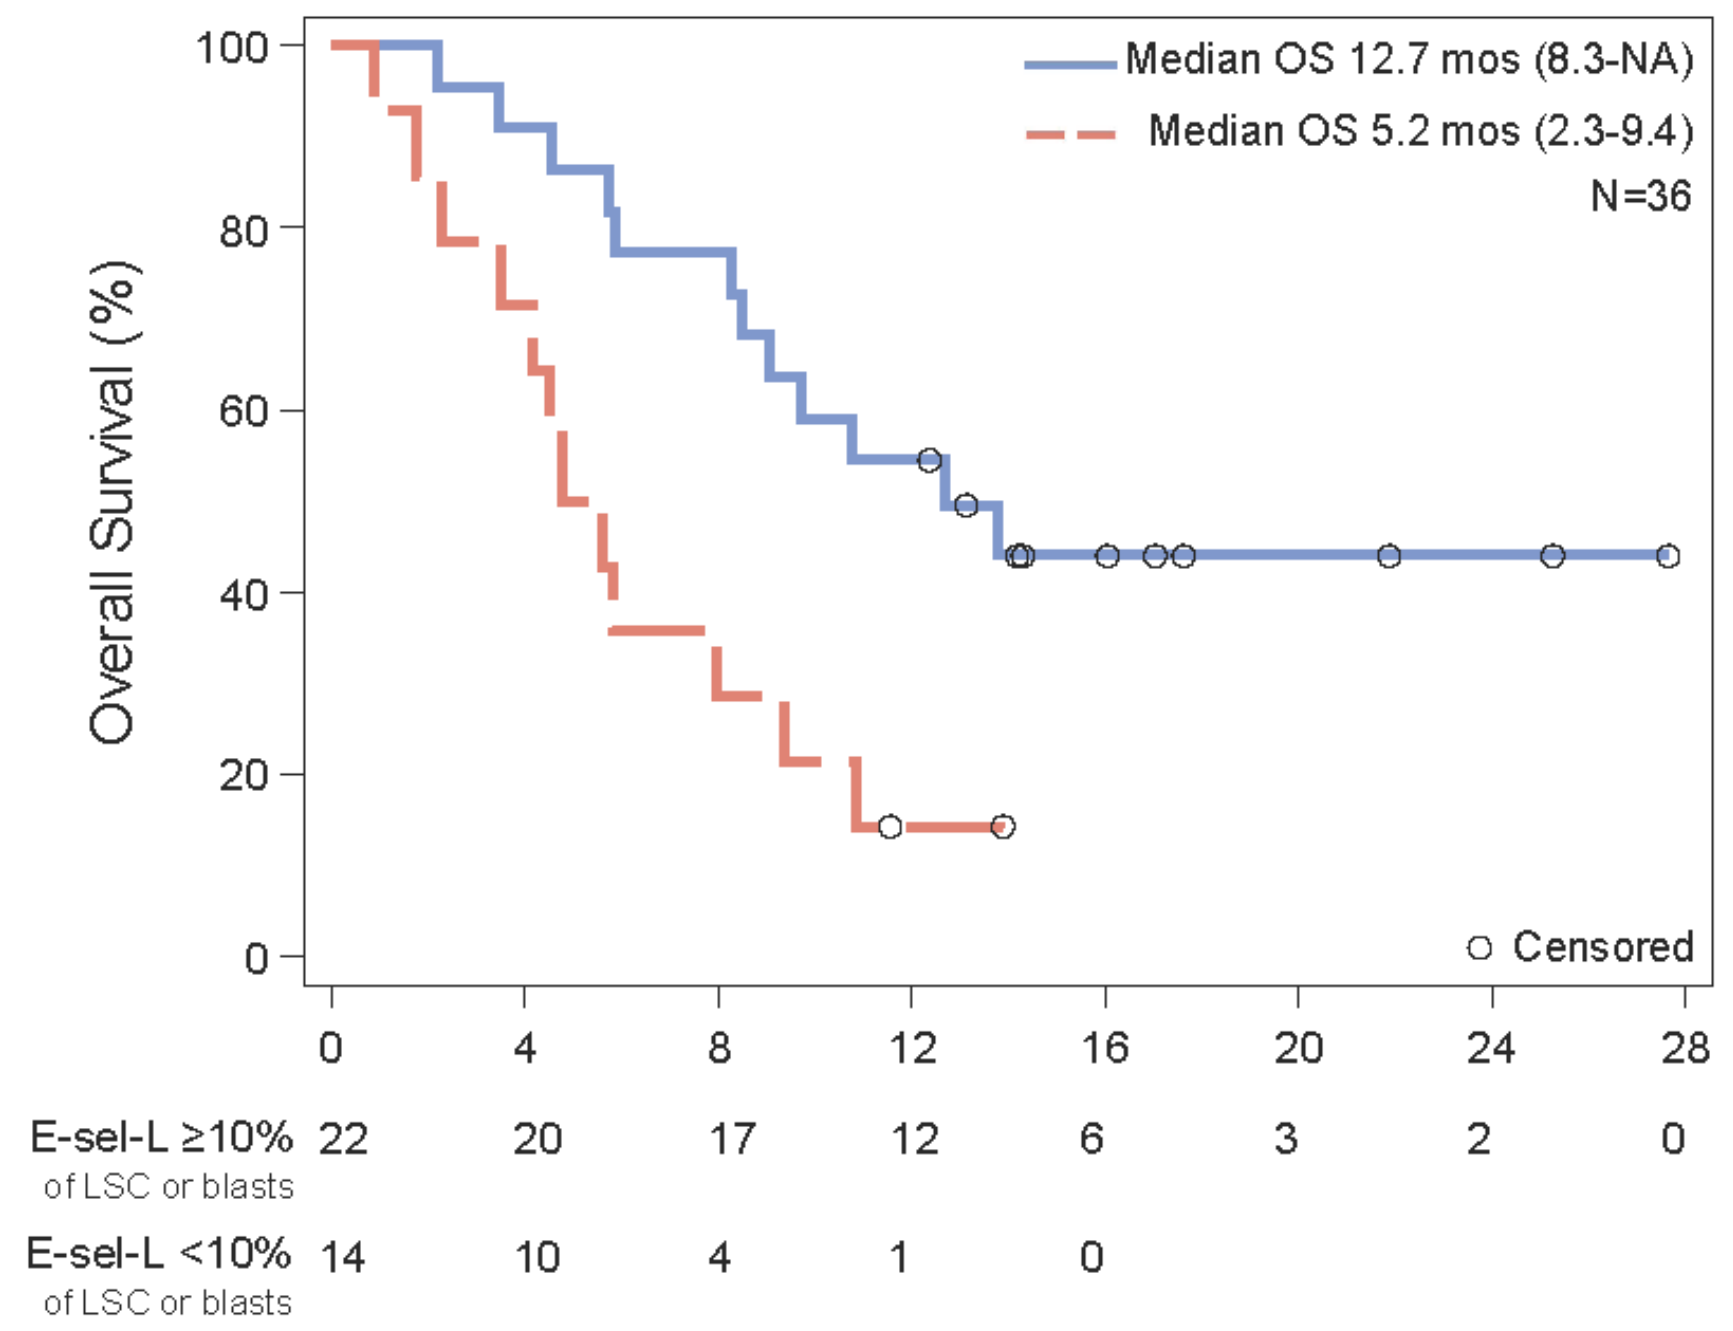

Median OS 12.7 mos (8.3-NA)
Median OS 5.2 mos (2.3-9.4)
N=36
Overall Survival (%)
Censored
E-sel-L ≥10% of LSC or blasts 22 20 17 12 6 3 2 0
E-sel-L <10% of LSC or blasts 14 10 4 1 0

Key Criteria for Study Entry

- ≥18 to ≤75 years old
- Primary refractory AML defined as follows:
 - Must have received 1 (and only 1) prior induction regimen containing both an anthracycline and cytarabine*‡
 - Persistent disease (≥5% blasts in bone marrow) at least 28 days after initiation of induction therapy
 - -- OR --
 - Relapse from a first remission [CR, CRi, CRp) lasting for <90 days
- Relapsed AML defined as follows:
 - First or second relapse untreated with cytotoxic regimen*‡
 - Secondary refractory AML is not allowed
- Prior transplant (HSCT) is allowed
 - HSCT >4 months
 - No acute GVHD ≥ Grade 2
 - No active chronic GVHD requiring immunosuppressive therapy
- Must be medically eligible for chemotherapy
- ECOG performance status ≤2

*Single agent target inhibitors are not considered cytotoxic chemotherapy for the purpose of the study:
- FLT3 inhibitors
- IDH1/IDH2 inhibitors

‡HMA +/- venetoclax

Uproleselan Relapsed/Refractory AML Phase 3 Trial Design

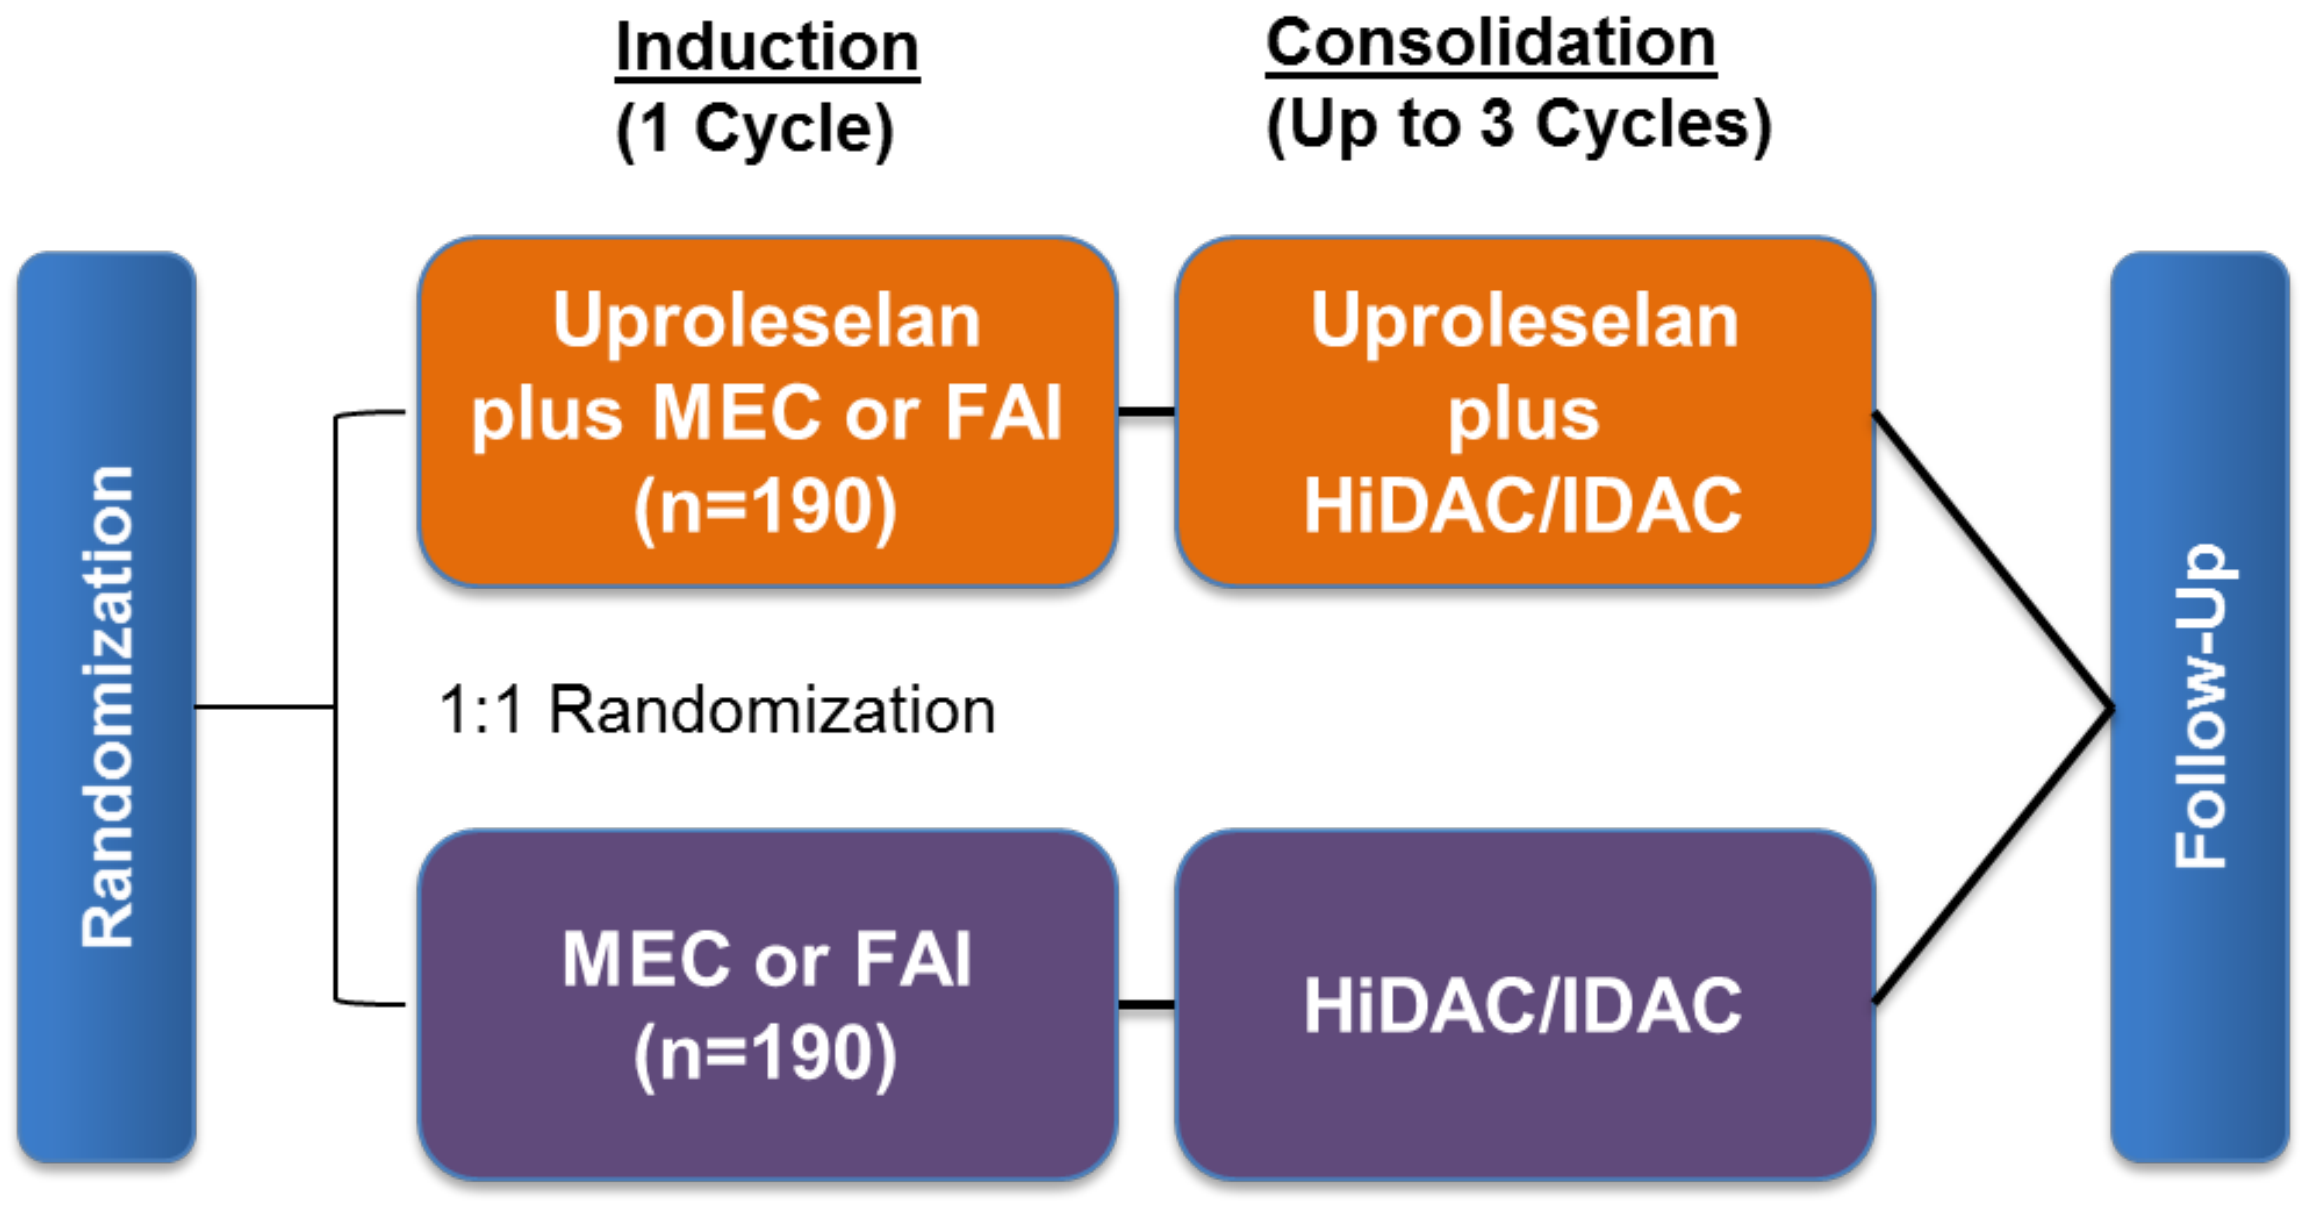

Induction
(1 Cycle)
Consolidation
(Up to 3 Cycles)
Randomization
Uproleselan plus MEC or FAI (n=190)
Uproleselan plus HiDAC/IDAC
1:1 Randomization
MEC or FAI (n=190)
HiDAC/IDAC
Follow-Up

After completion of the induction period and response assessment, every effort should be made to transplant eligible subjects. For those in remission and being actively assessed for transplant, consolidation on trial while awaiting transplant is strongly encouraged.

MEC: Mitoxantrone, etoposide and cytarabine; FAI: Fludarabine, cytarabine and idarubicin; HiDAC/IDAC: High-dose or Intermediate-dose cytarabine; based on investigator's choice

Treatment

Uproleselan/placebo treatment:
- Pre-chemotherapy: 24 hours prior to first dose of chemotherapy at 800 mg
- During chemotherapy: 800 mg Q12h x 5 days
- Post-chemotherapy: 800 mg Q12h x 2 days

Patients will receive the single priming dose of uproleselan/placebo at 800 mg given 24 hours prior to first dose of chemotherapy, every 12 hours (± 1 hour) on chemotherapy days and 2 days following the last dose of chemotherapy

Current Enrollment

This trial is expected to enroll 380 patients across approximately 8 countries in North America, Europe, and Australia (Figure 5). The first patient was enrolled in November 2018.

Figure 5: Participating Countries

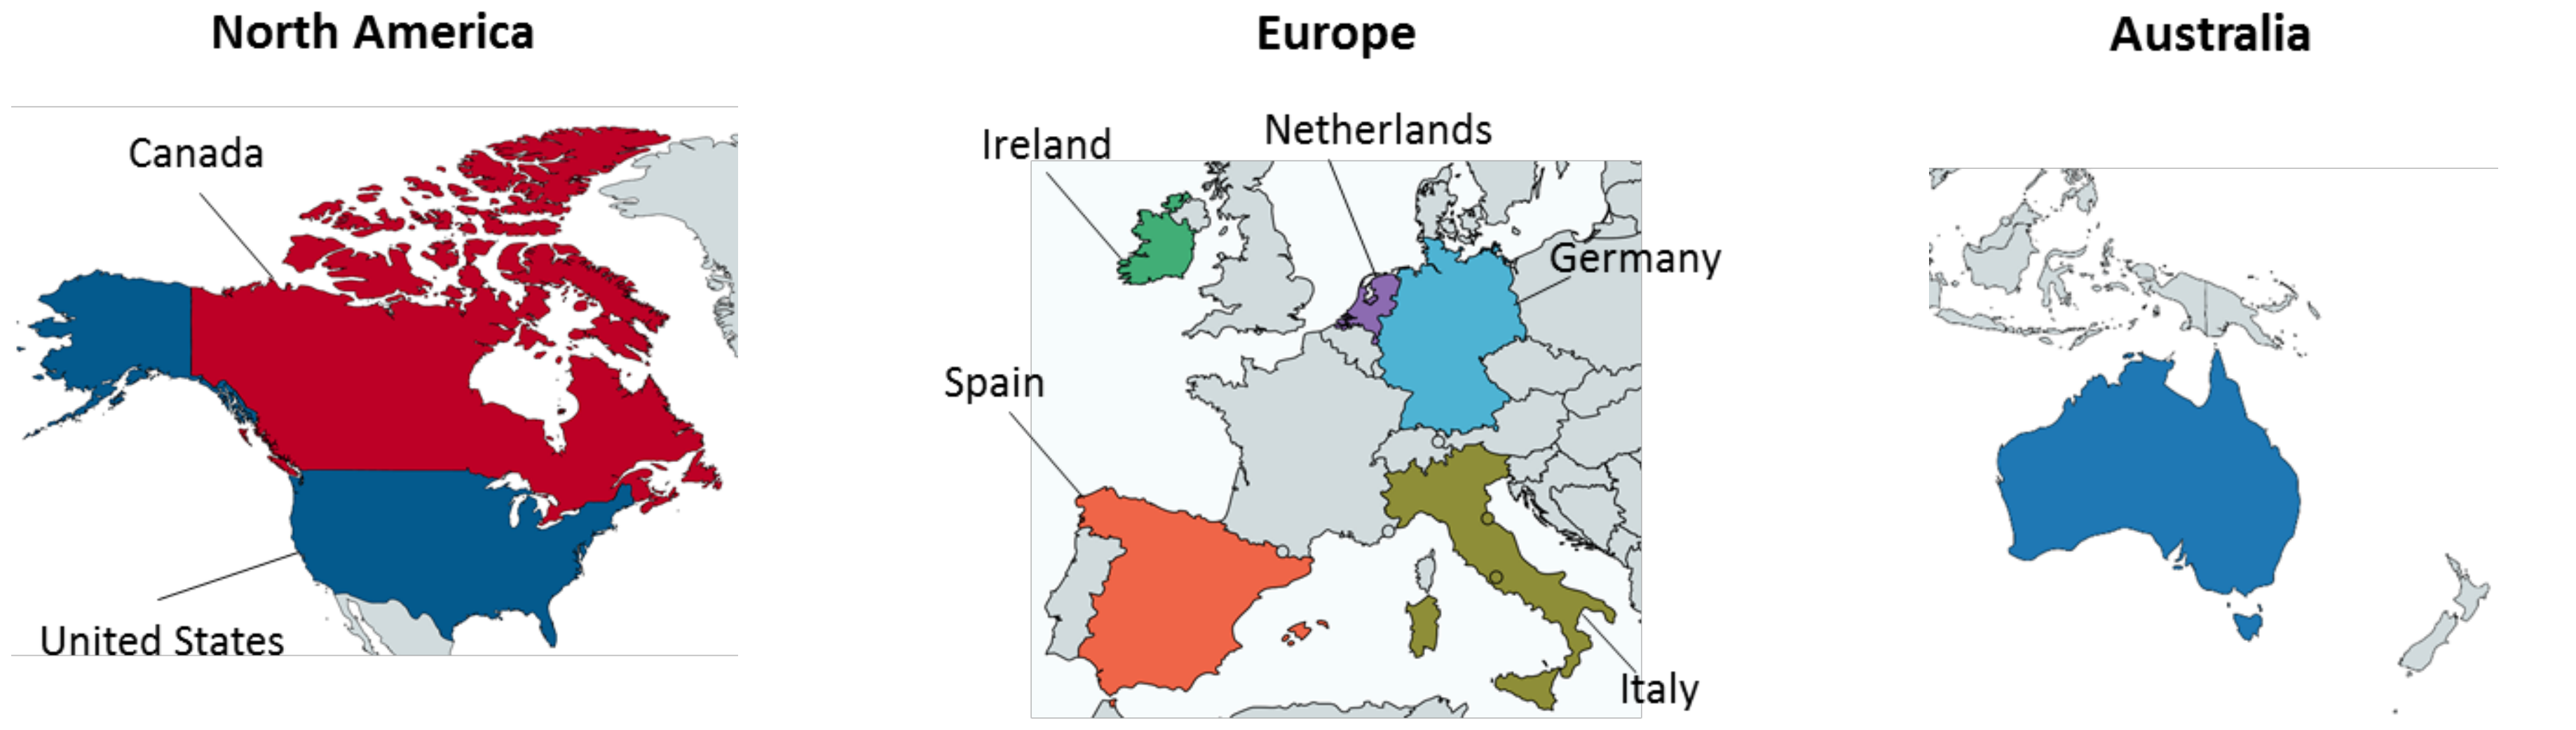

North America
Europe
Australia
Canada
United States
Ireland
Netherlands
Germany
Spain
Italy

Endpoints

Primary Endpoint:
- To evaluate OS achieved with uproleselan administered with chemotherapy versus chemotherapy alone

Key secondary endpoints
- Rate of severe oral mucositis
- Rate of HSCT
- Rate of CR and CRh

Exploratory endpoints
- MRD assessment
- E-selectin ligand expression on leukemic blast cells, plasma soluble E-selectin

References

DeAngelo DJ, et al. Blood 2018; 132:331; DeAngelo DJ, et al. Blood 2017; 130:894; Feldman E, et al. J Clin Oncol 2005; 23 (18):4110; Angelini DE, et al. Blood 2016;128:3826; Cheson BD, et al. J Clin Oncol 2003;21 (24):4642-4649; Chien S, et al. Blood 2013;122:2161; Chien S, et al. Blood 2012;120:4092; Devata S, et al. Blood 2015;126:1004; Dohner H, et al. Blood 2010;115 (3):453-474; Rashidi A, DiPersio JF. Ther Adv Hematol 2016;7:4051; Winkler IG, et al. Nat Med 2012;18(11):1651-7; Winkler IG, et al. Blood 2013;122:2266; Winkler IG, et al. Blood 2014;124:620; Winkler IG, et al. Blood 2016; 128:2823.

Conflict of Interest Disclosures: DJD, HE, PM, GH, BC, BB, and MA have no conflicts to report. MEO, JLL, and PSB received research funding, and BAJ and ASA were paid consultants over the past 2 years for GlycoMimetics, Inc. WF, MC, JM, HT, and EF are employees of, and have equity ownership in, GlycoMimetics, Inc.
Funding of this trial : GlycoMimetics, Inc.
ClinicalTrials.gov Identifier: NCT03616470

For further information please contact efeldman@glycomimetics.com

More information on this and related GlycoMimetics projects can be obtained at glycomimetics.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster